CBIO-14

Targeting Proteasome Activity with Marizomib as a Therapeutic Perspective for Glioma Patients

Shahrzad Jalali[1], Ann MacLaren[2], Francis Burrows[2], H. Artee Luchman[3], Mohit Trikha[2], Gelareh Zadeh[1] and Kenneth Aldape[1]

[1] Princess Margaret Cancer Center, University of Toronto, ON, Canada; [2] Triphase Accelerator, ON, Canada & CA, USA, [3]Hotchkiss Brain Institute, University of Calgary, AB, Canada

Abstract

Inhibition of the Ubiquitin-Proteasome pathway offers promise for the treatment of gliomas, however, this treatment strategy is unproven as the approved drugs in this class do not penetrate the CNS. Marizomib is a novel second-generation proteasome inhibitor, with advantages over bortezomib and carfilzomib, including irreversible inhibition of all three enzymatic activities of the proteasome and superior tolerability. While there are some promising data in preclinical models of solid tumors using proteasome inhibitors, marizomib has also proven to be active in intracranial glioma xenografts, prompting the initiation of a Phase I trial in glioblastoma (GB) in combination with bevacizumab (BEV). We aim to identify potential biomarkers that would enable prediction of response to marizomib in GB patients.

Tumor tissues obtained from glioma patients at Toronto Western Hospital were utilized for proteasome activity assays.

Results demonstrate a significant increase in all proteasome activities, chymotrypsin-like (CT-L), trypsin-like (T-L), and caspase-like (C-L), in GB tumors compared to normal brain tissues. Furthermore, progression of low grade astrocytoma to GB is also associated with enhanced CT-L and T-L activities, indicating that targeting these subunits could potentially inhibit the progression of gliomas.

In conclusion, marizomib represents a potential therapeutic agent for GB. Further investigation is necessary to assess the therapeutic benefits of marizomib as a single agent or in combination with other agents.

Results

Differential sensitivity to proteasome inhibition in patient derived glioma cells

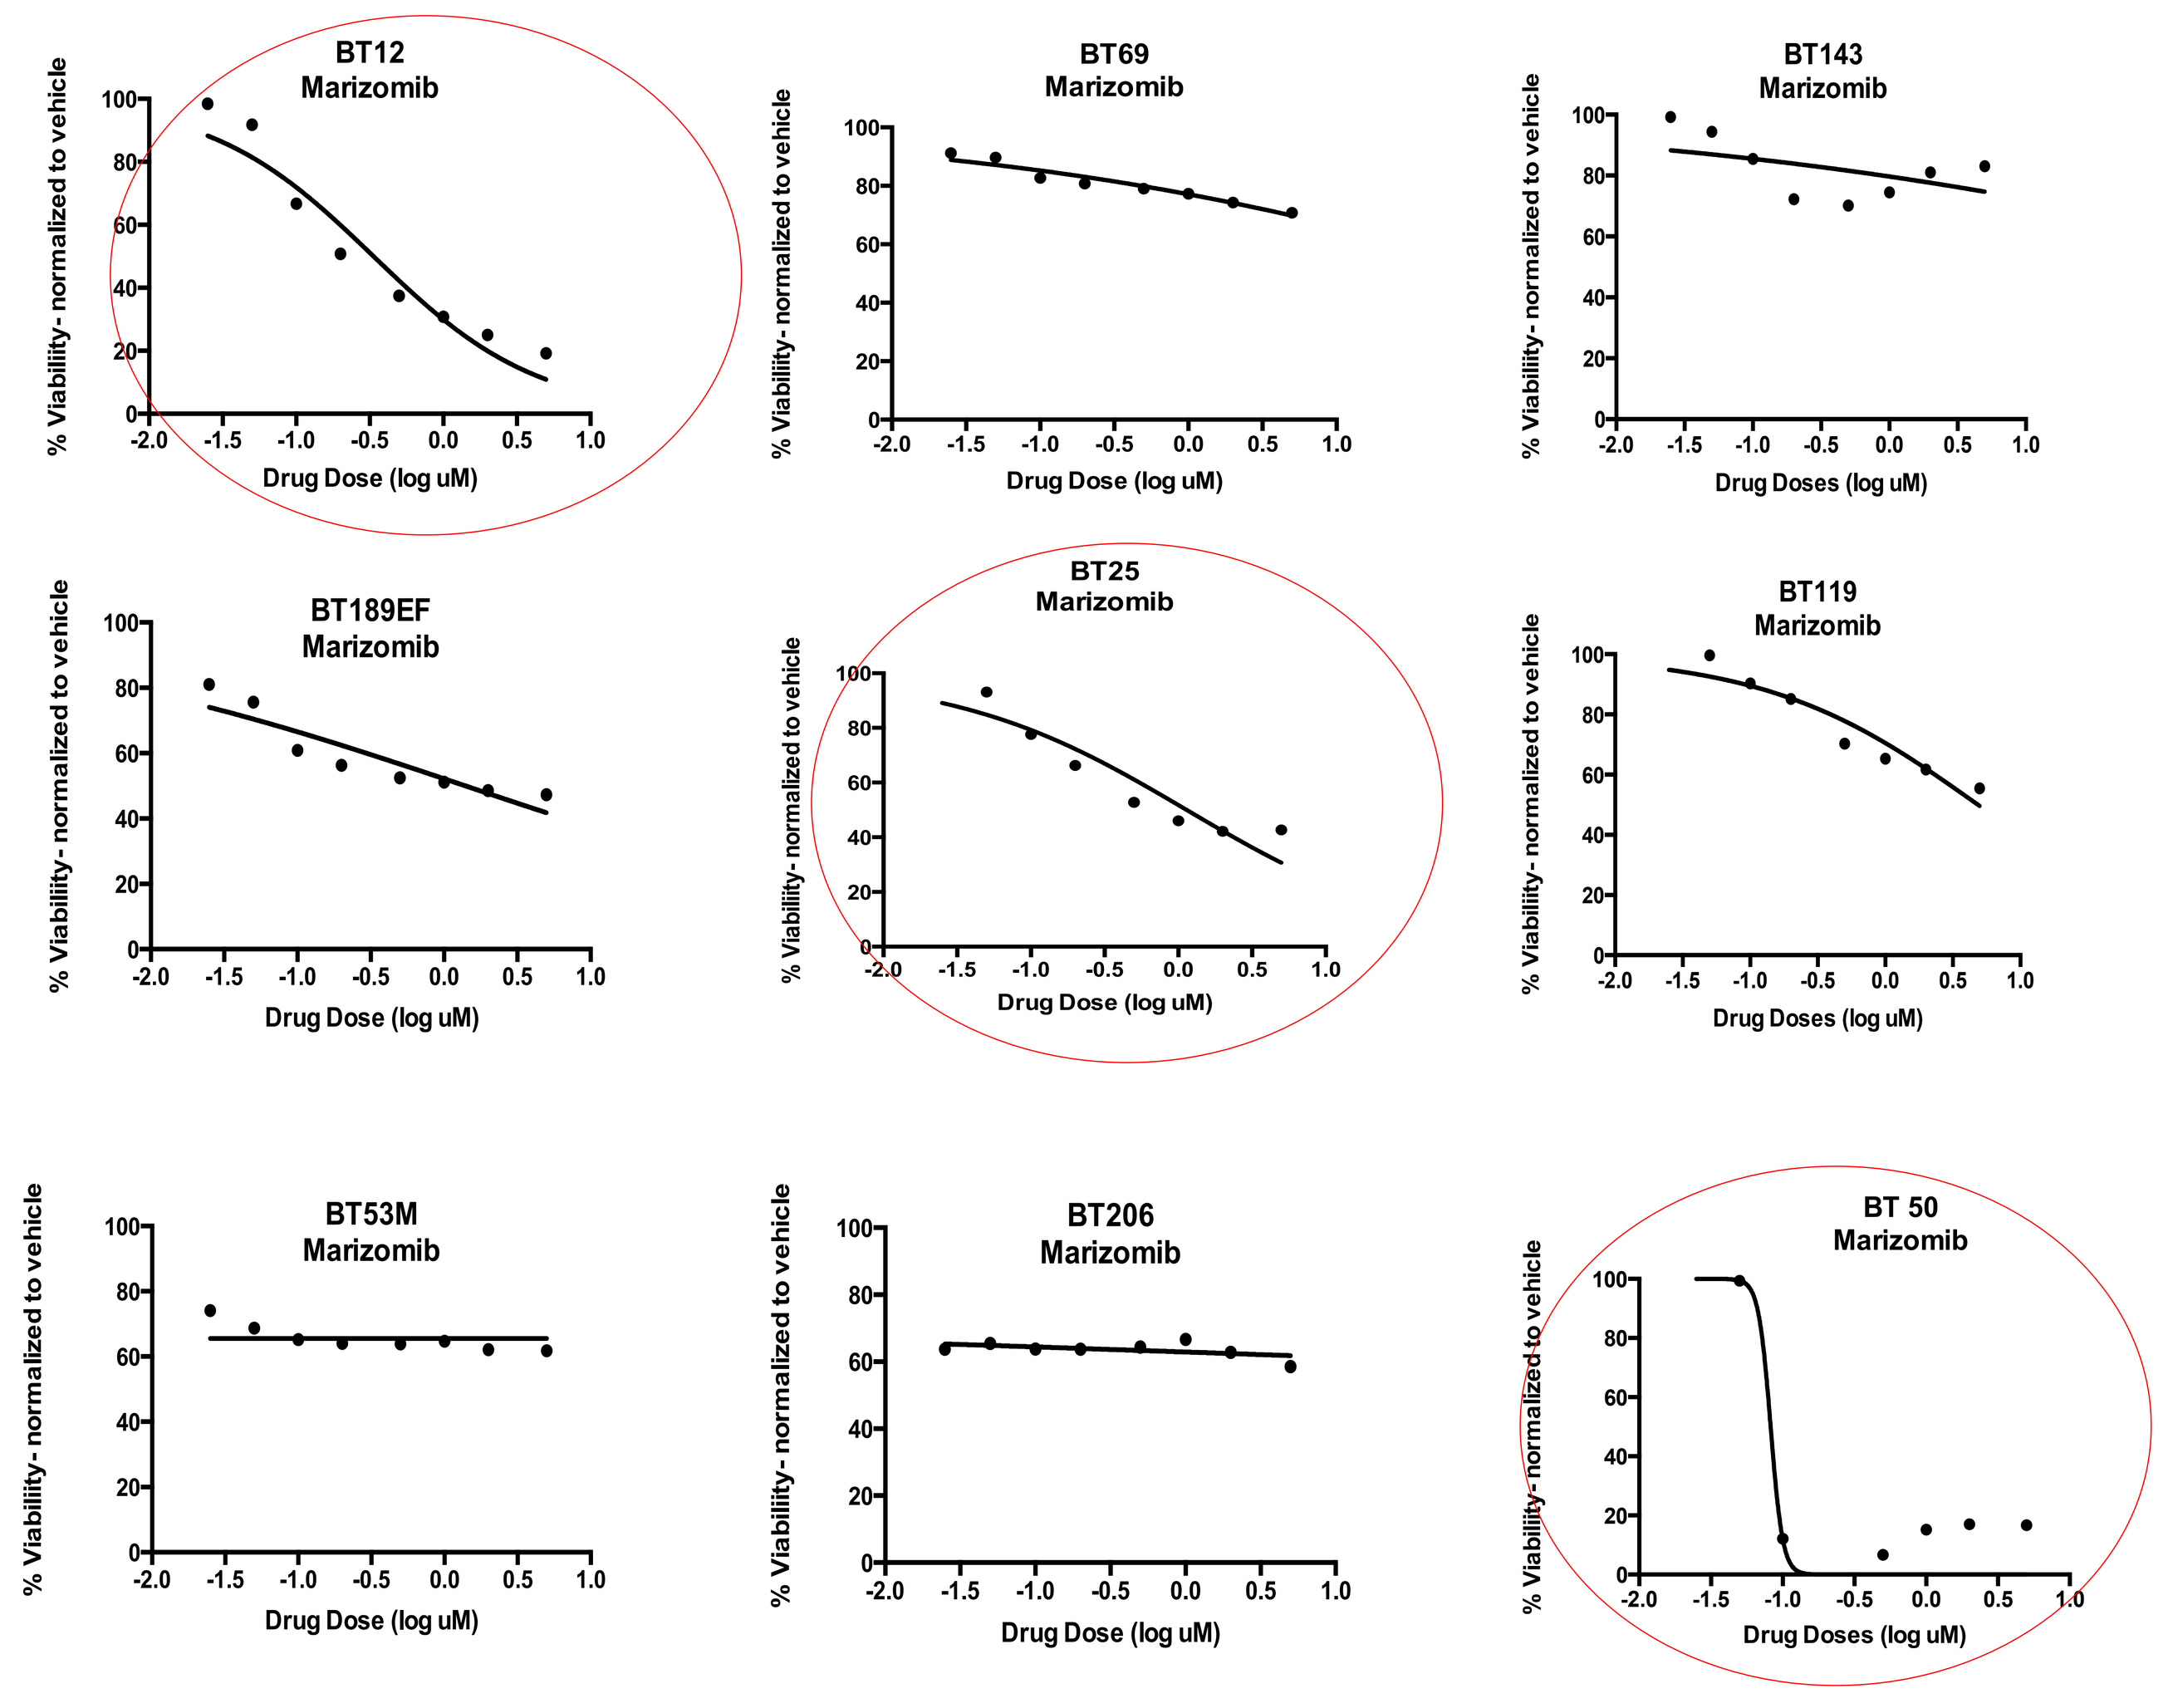
BT12 Marizomib
BT69 Marizomib
BT143 Marizomib
BT189EF Marizomib
BT25 Marizomib
BT119 Marizomib
BT53M Marizomib
BT206 Marizomib
BT 50 Marizomib
% Viability- normalized to vehicle
Drug Dose (log uM)
Drug Doses (log uM)

- **Glioma stem cells (GSCs) have been demonstrated to drive tumor initiation and have similar drug responses to human gliomas**
- **Marizomib treatment in a panel of 9 GSCs demonstrated that ~30% of these cell lines show unique sensitivity to proteasome inhibition (red circles)**
- **These data suggest that proteasome inhibition may have a variable response in the glioma population and that a predictive biomarker strategy will likely be required to stratify patients who might best respond to MRZ treatment**

Proteasome activity is elevated in patient derived glioblastoma samples

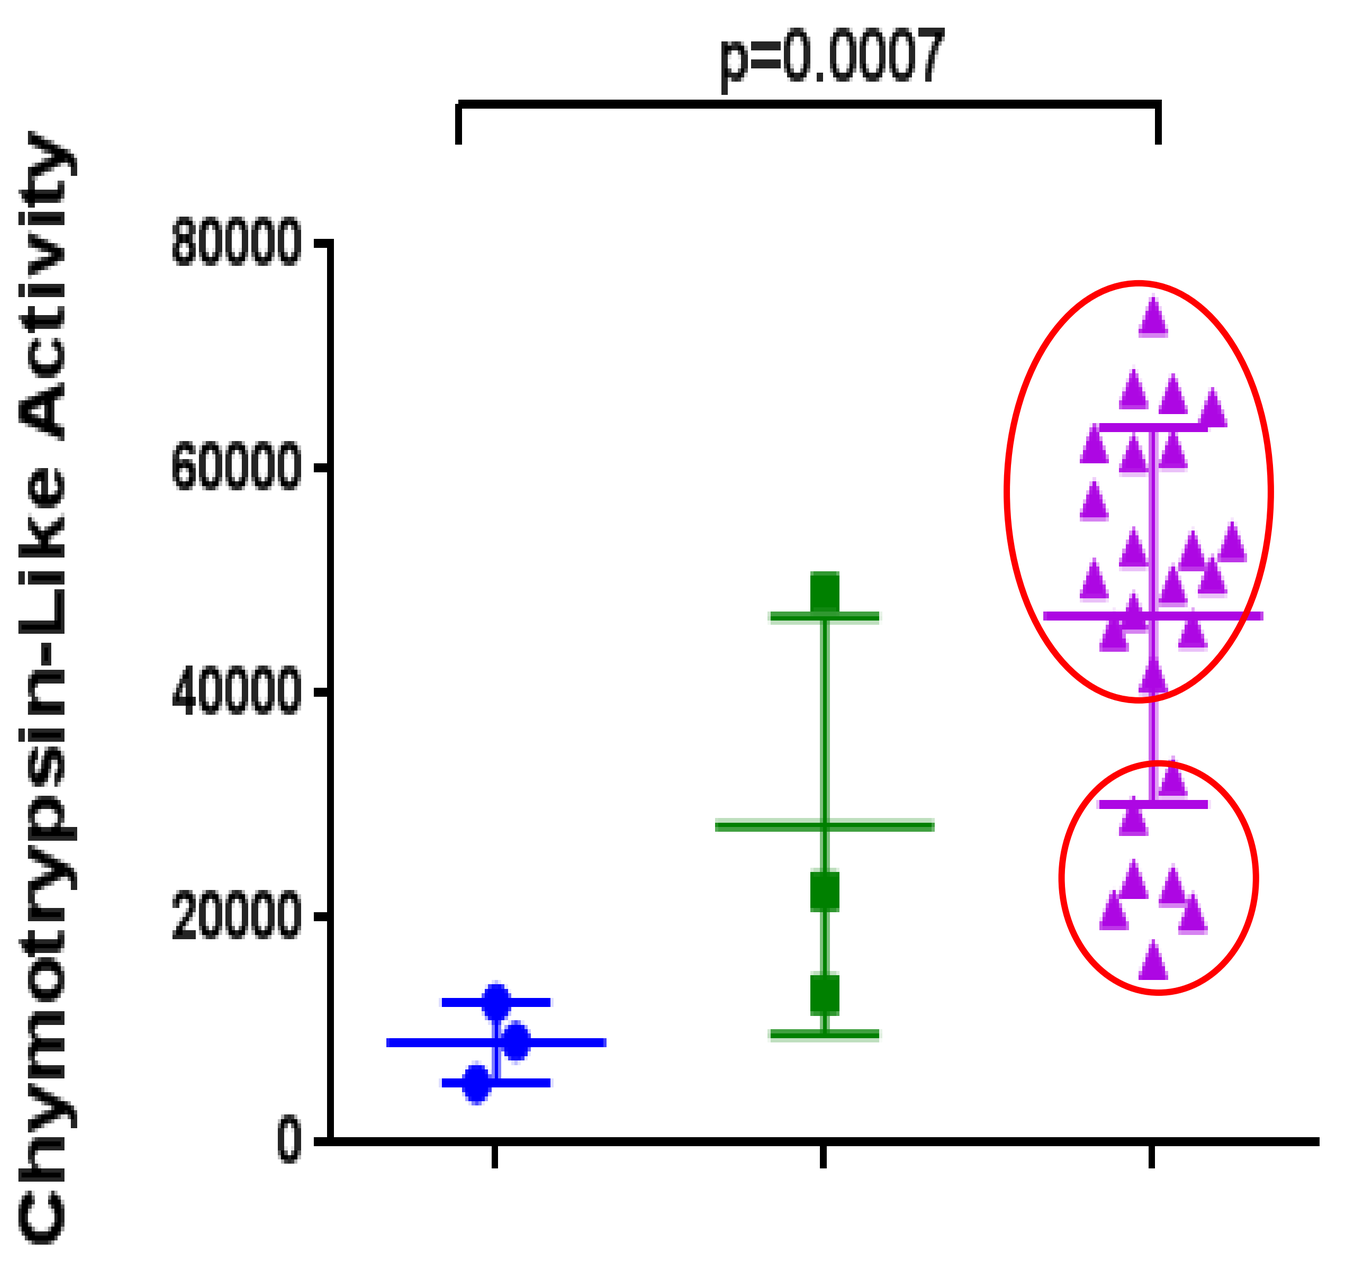
p=0.0007
Chymotrypsin-Like Activity
80000
60000
40000
20000
0

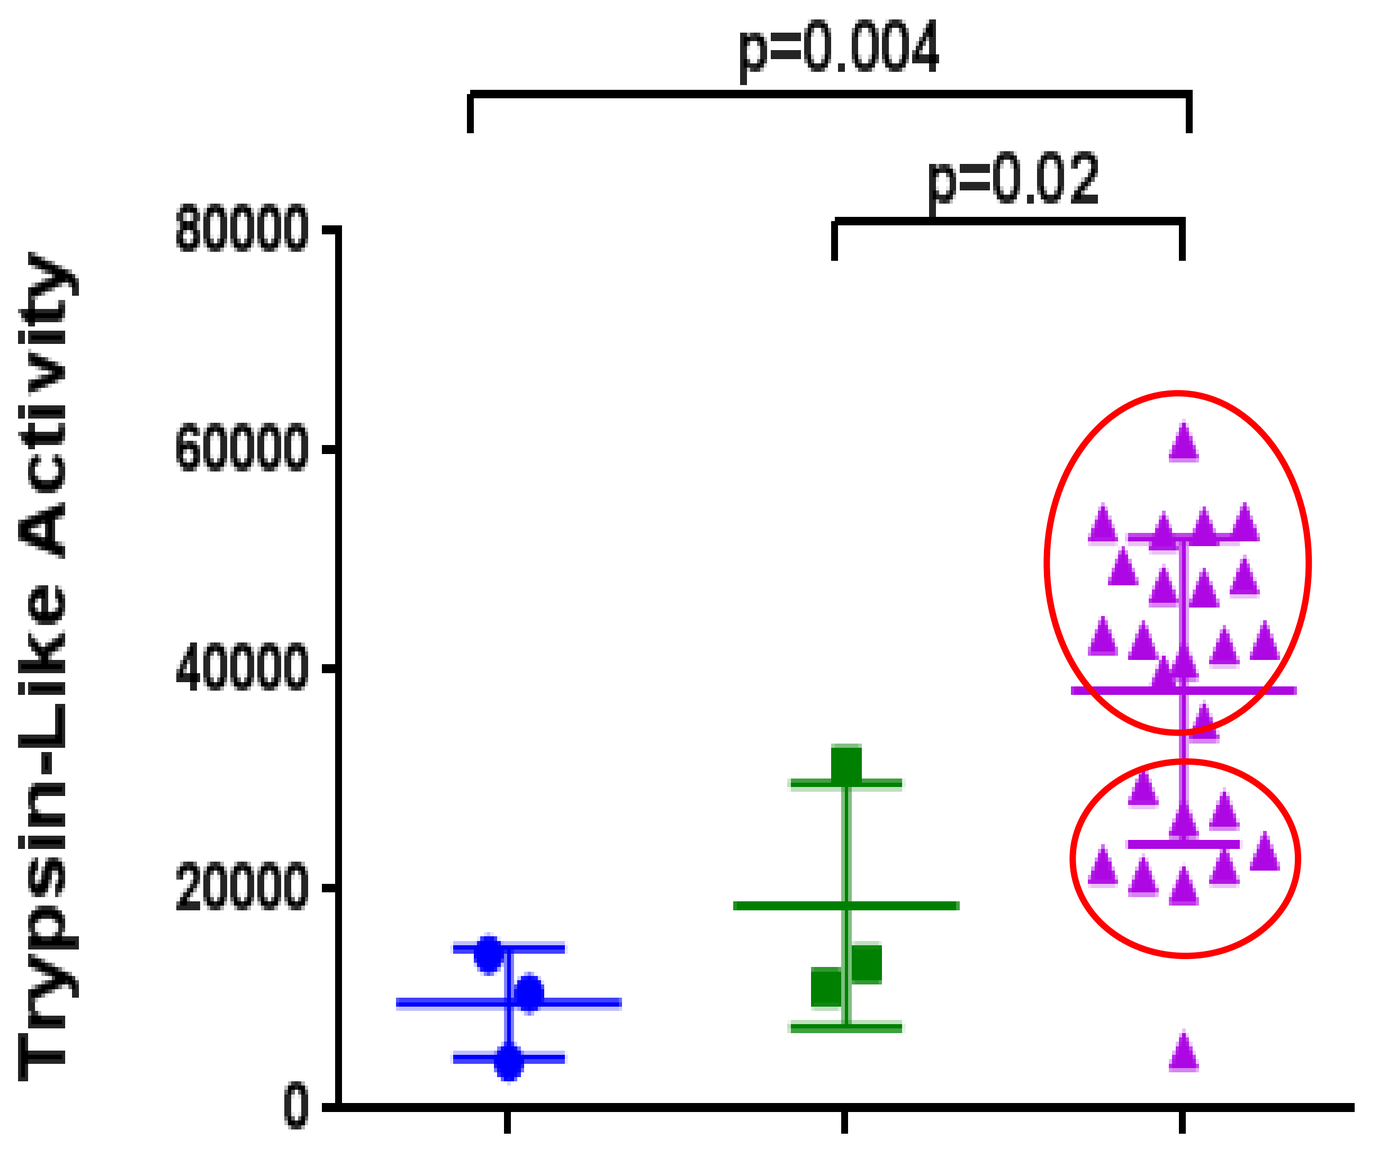
p=0.004
p=0.02
Trypsin-Like Activity
80000
60000
40000
20000
0

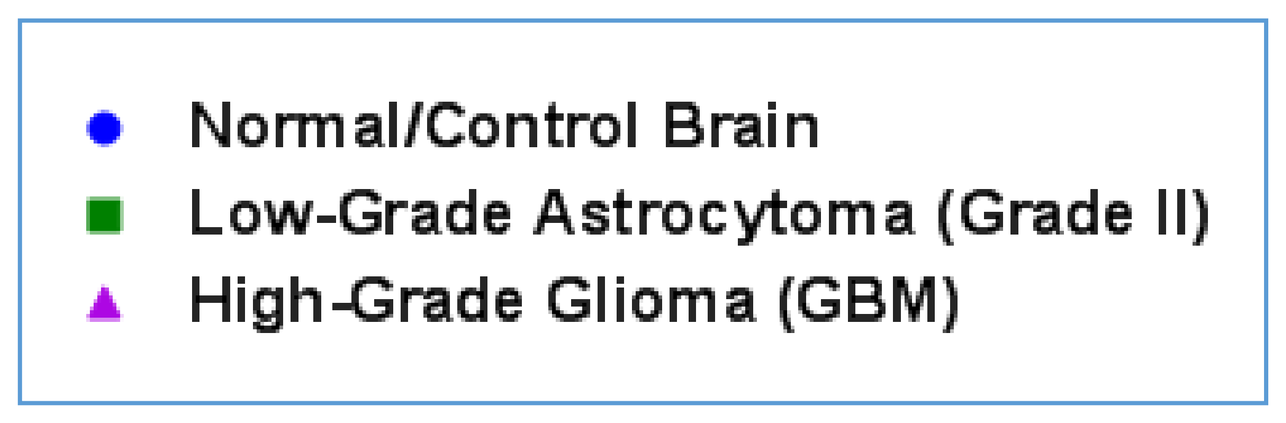
Normal/Control Brain
Low-Grade Astrocytoma (Grade II)
High-Grade Glioma (GBM)

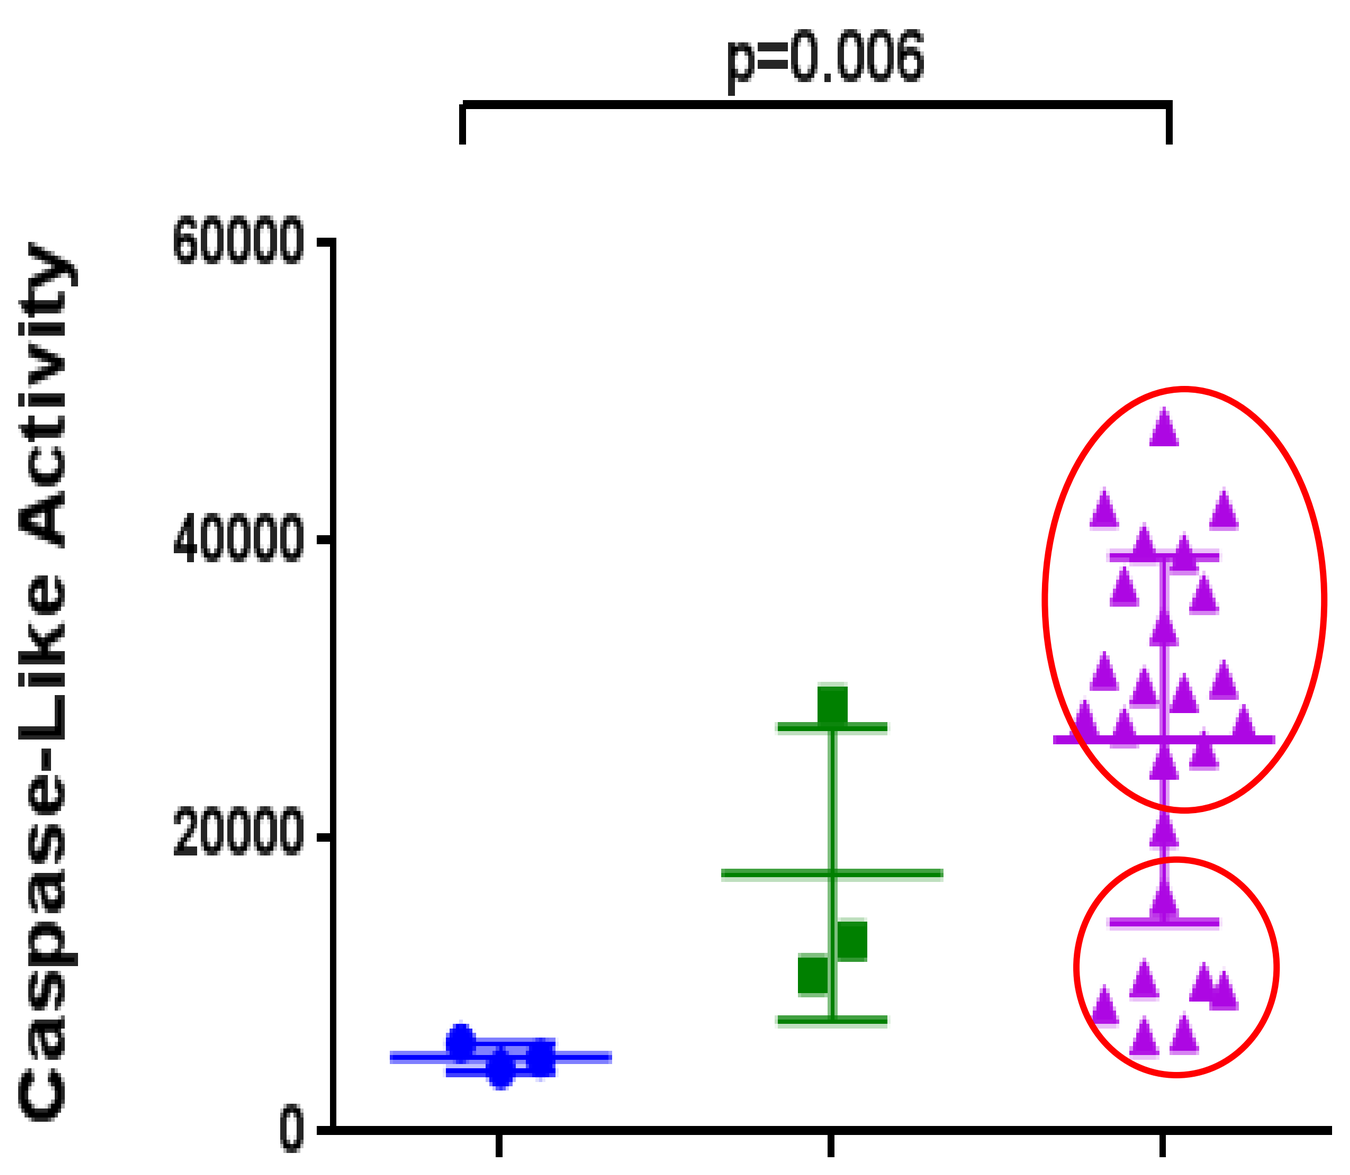
p=0.006
Caspase-Like Activity
60000
40000
20000
0

- **All three proteasome activities are elevated in high grade glioma**
- **Proteasome activity is elevated relative to normal brain and low grade astrocytoma**
- **Two subpopulations of proteasome activity exist in glioma; high and low proteasome activities were identified in high grade glioma patient lysates (circled in red)**

Strong correlation between CT-L & C-L proteasome activities in glioblastoma samples

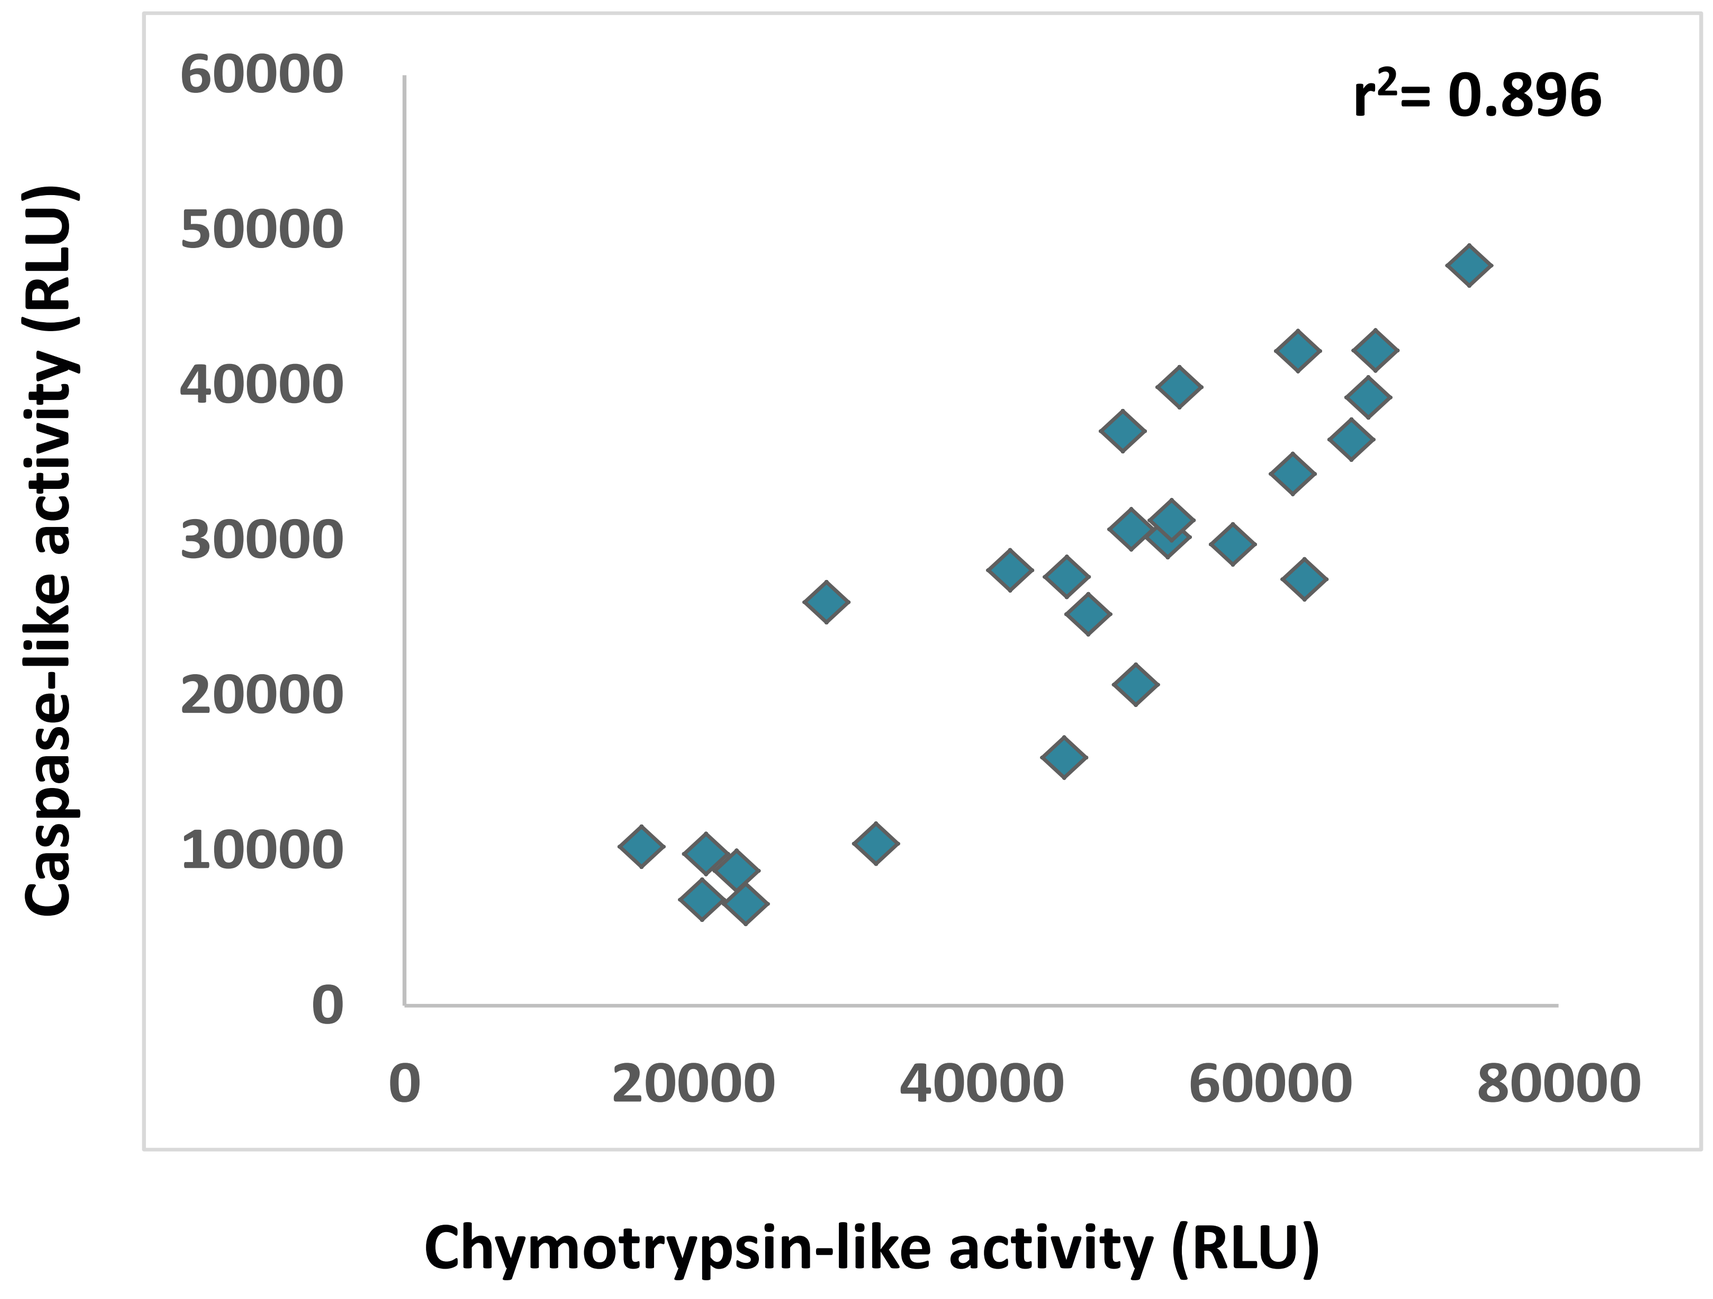
r^2= 0.896
Caspase-like activity (RLU)
Chymotrypsin-like activity (RLU)

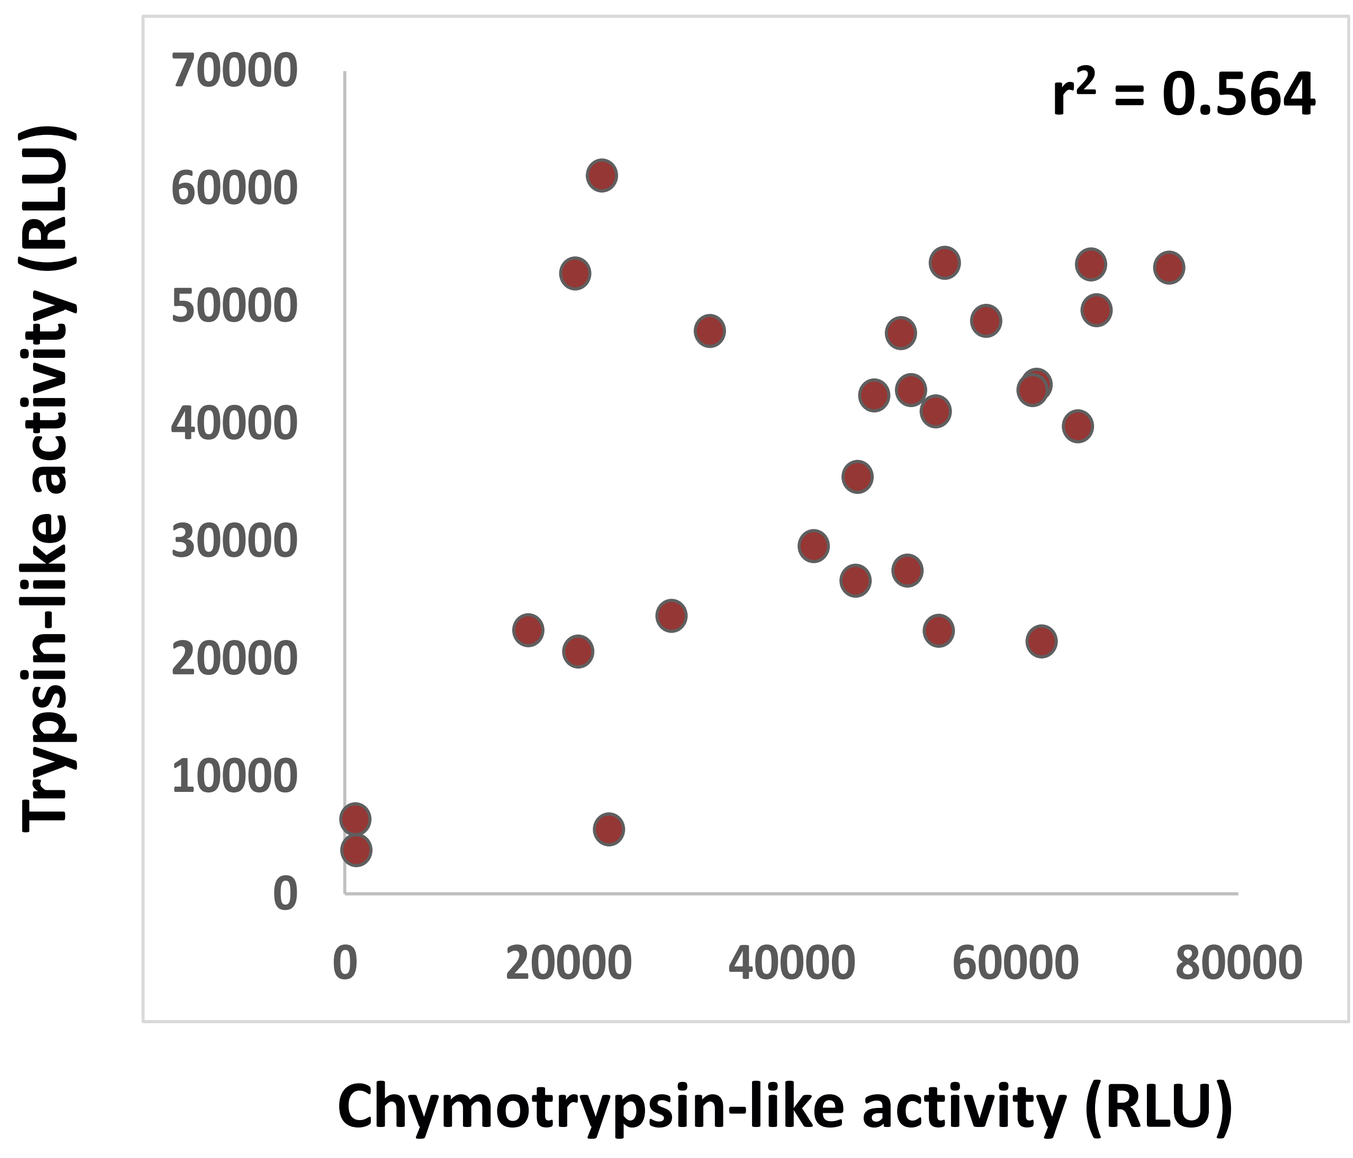
r^2 = 0.564
Trypsin-like activity (RLU)
Chymotrypsin-like activity (RLU)

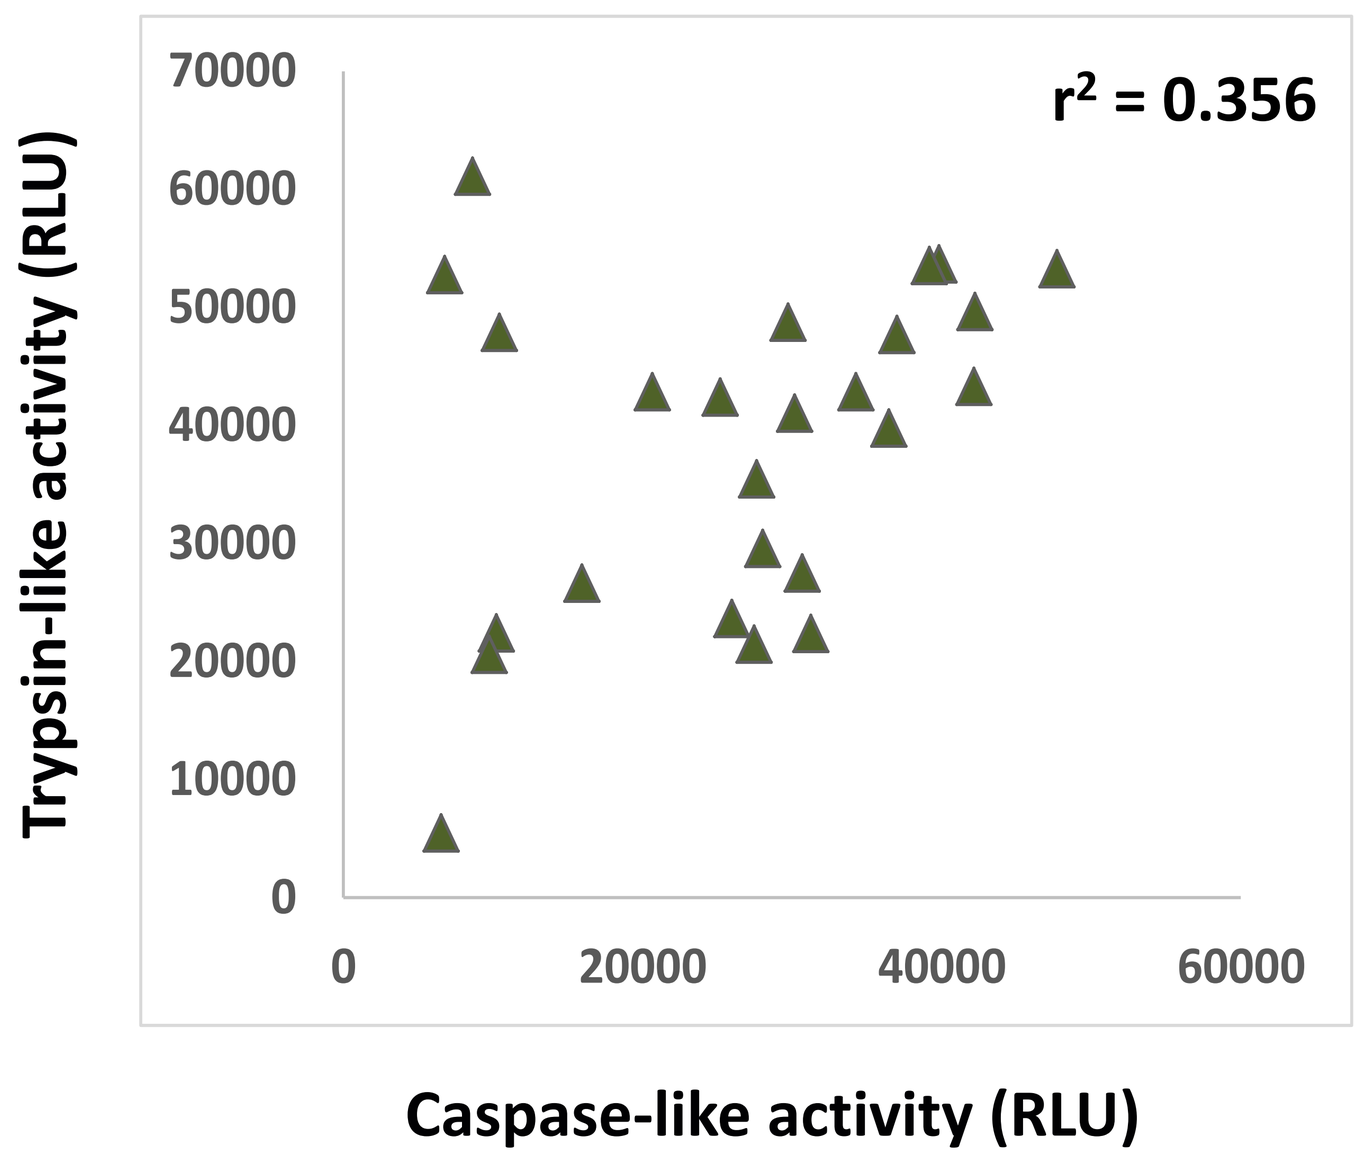
r^2 = 0.356
Trypsin-like activity (RLU)
Caspase-like activity (RLU)

- **A comparison of proteasome activity in high grade gliomas revealed a strong linear correlation between CT-L and C-L activities**
- **Patient samples with high CT-L activity, had a trend to higher C-L activity**
- **Less correlation was observed between C-L and T-L activities**

Proteasome activity increases with progression from WHO grade III to grade IV recurrent glioma

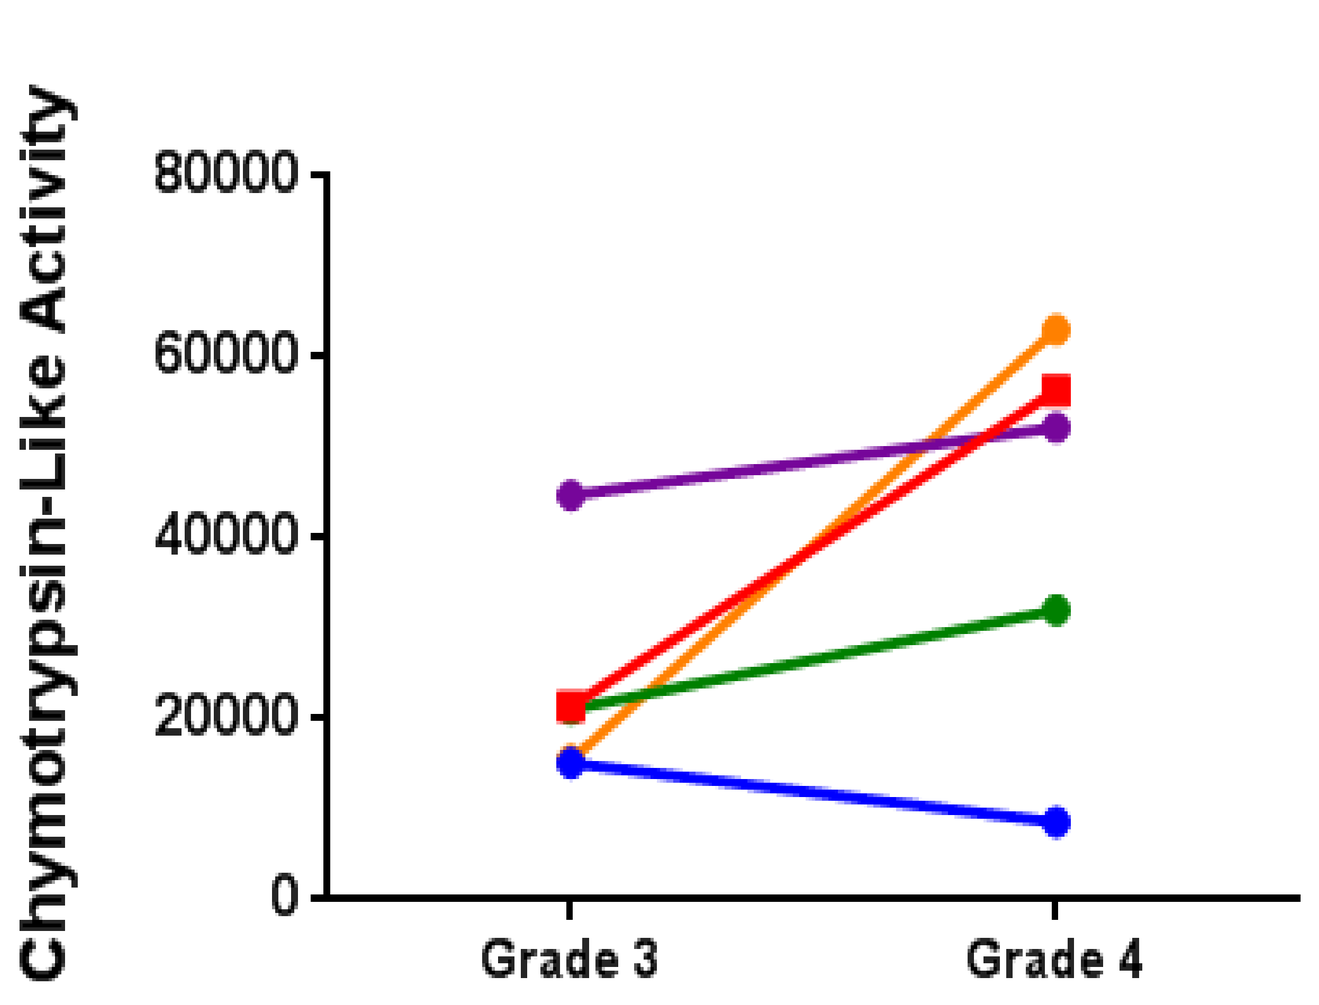
Chymotrypsin-Like Activity
Grade 3
Grade 4

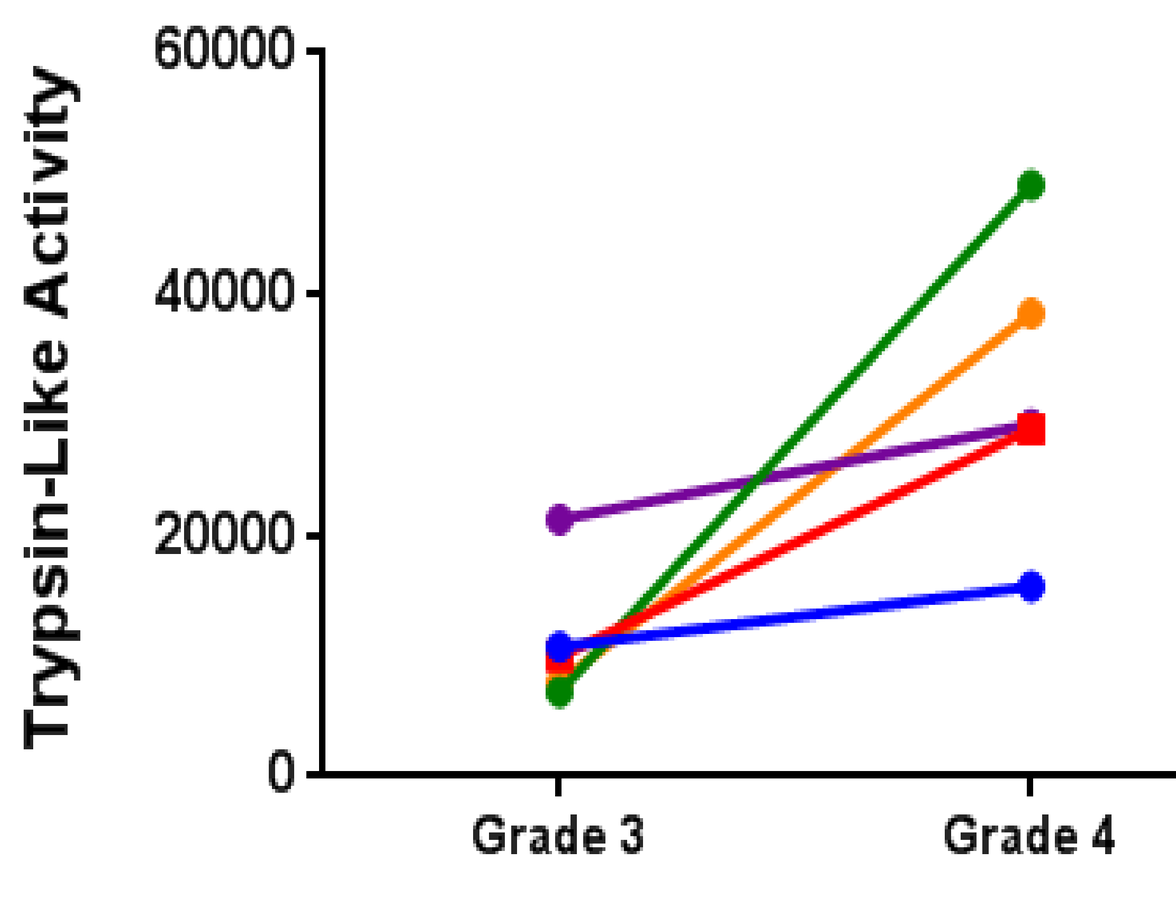
Trypsin-Like Activity
Grade 3
Grade 4

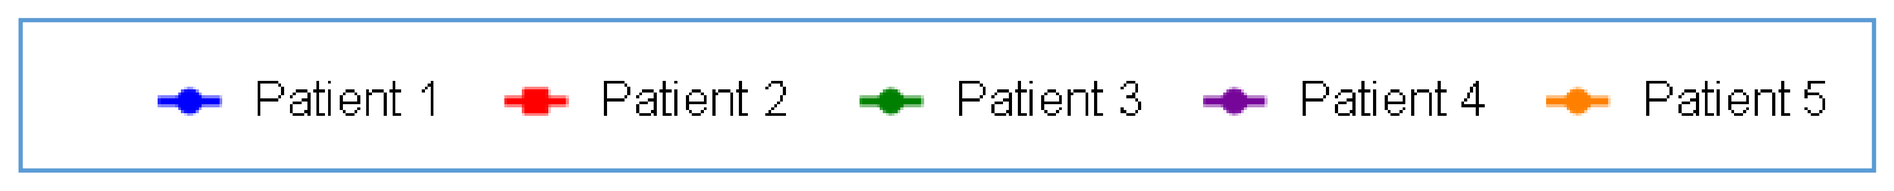
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5

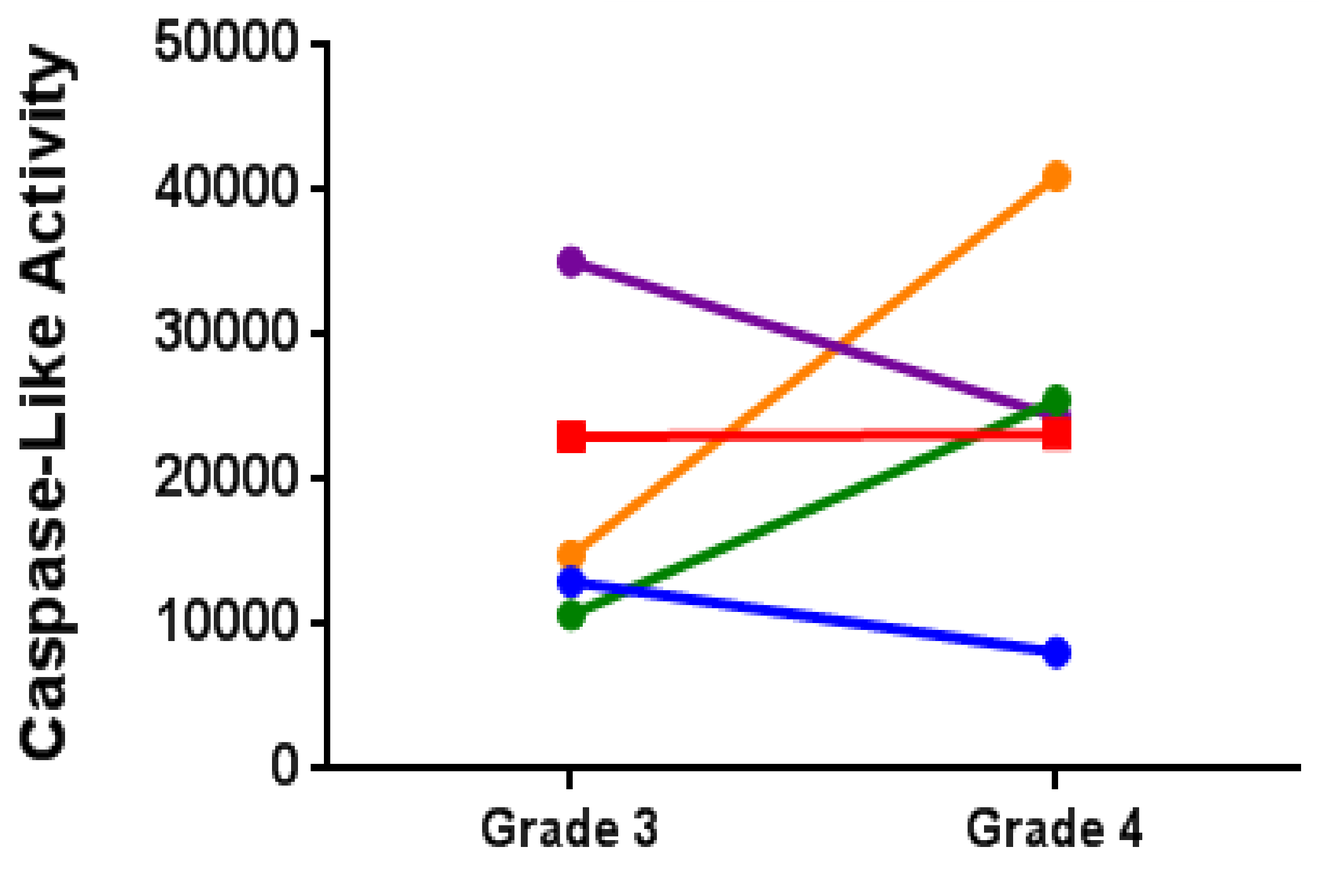
Caspase-Like Activity
Grade 3
Grade 4

- **Proteasome activities measured in 5 patients who progressed from grade III to grade IV glioma**
- **CT-L and T-L activities were increased with progression to high grade glioma**

Proteasome activity is elevated in primary and recurrent GBM compared to normal brain

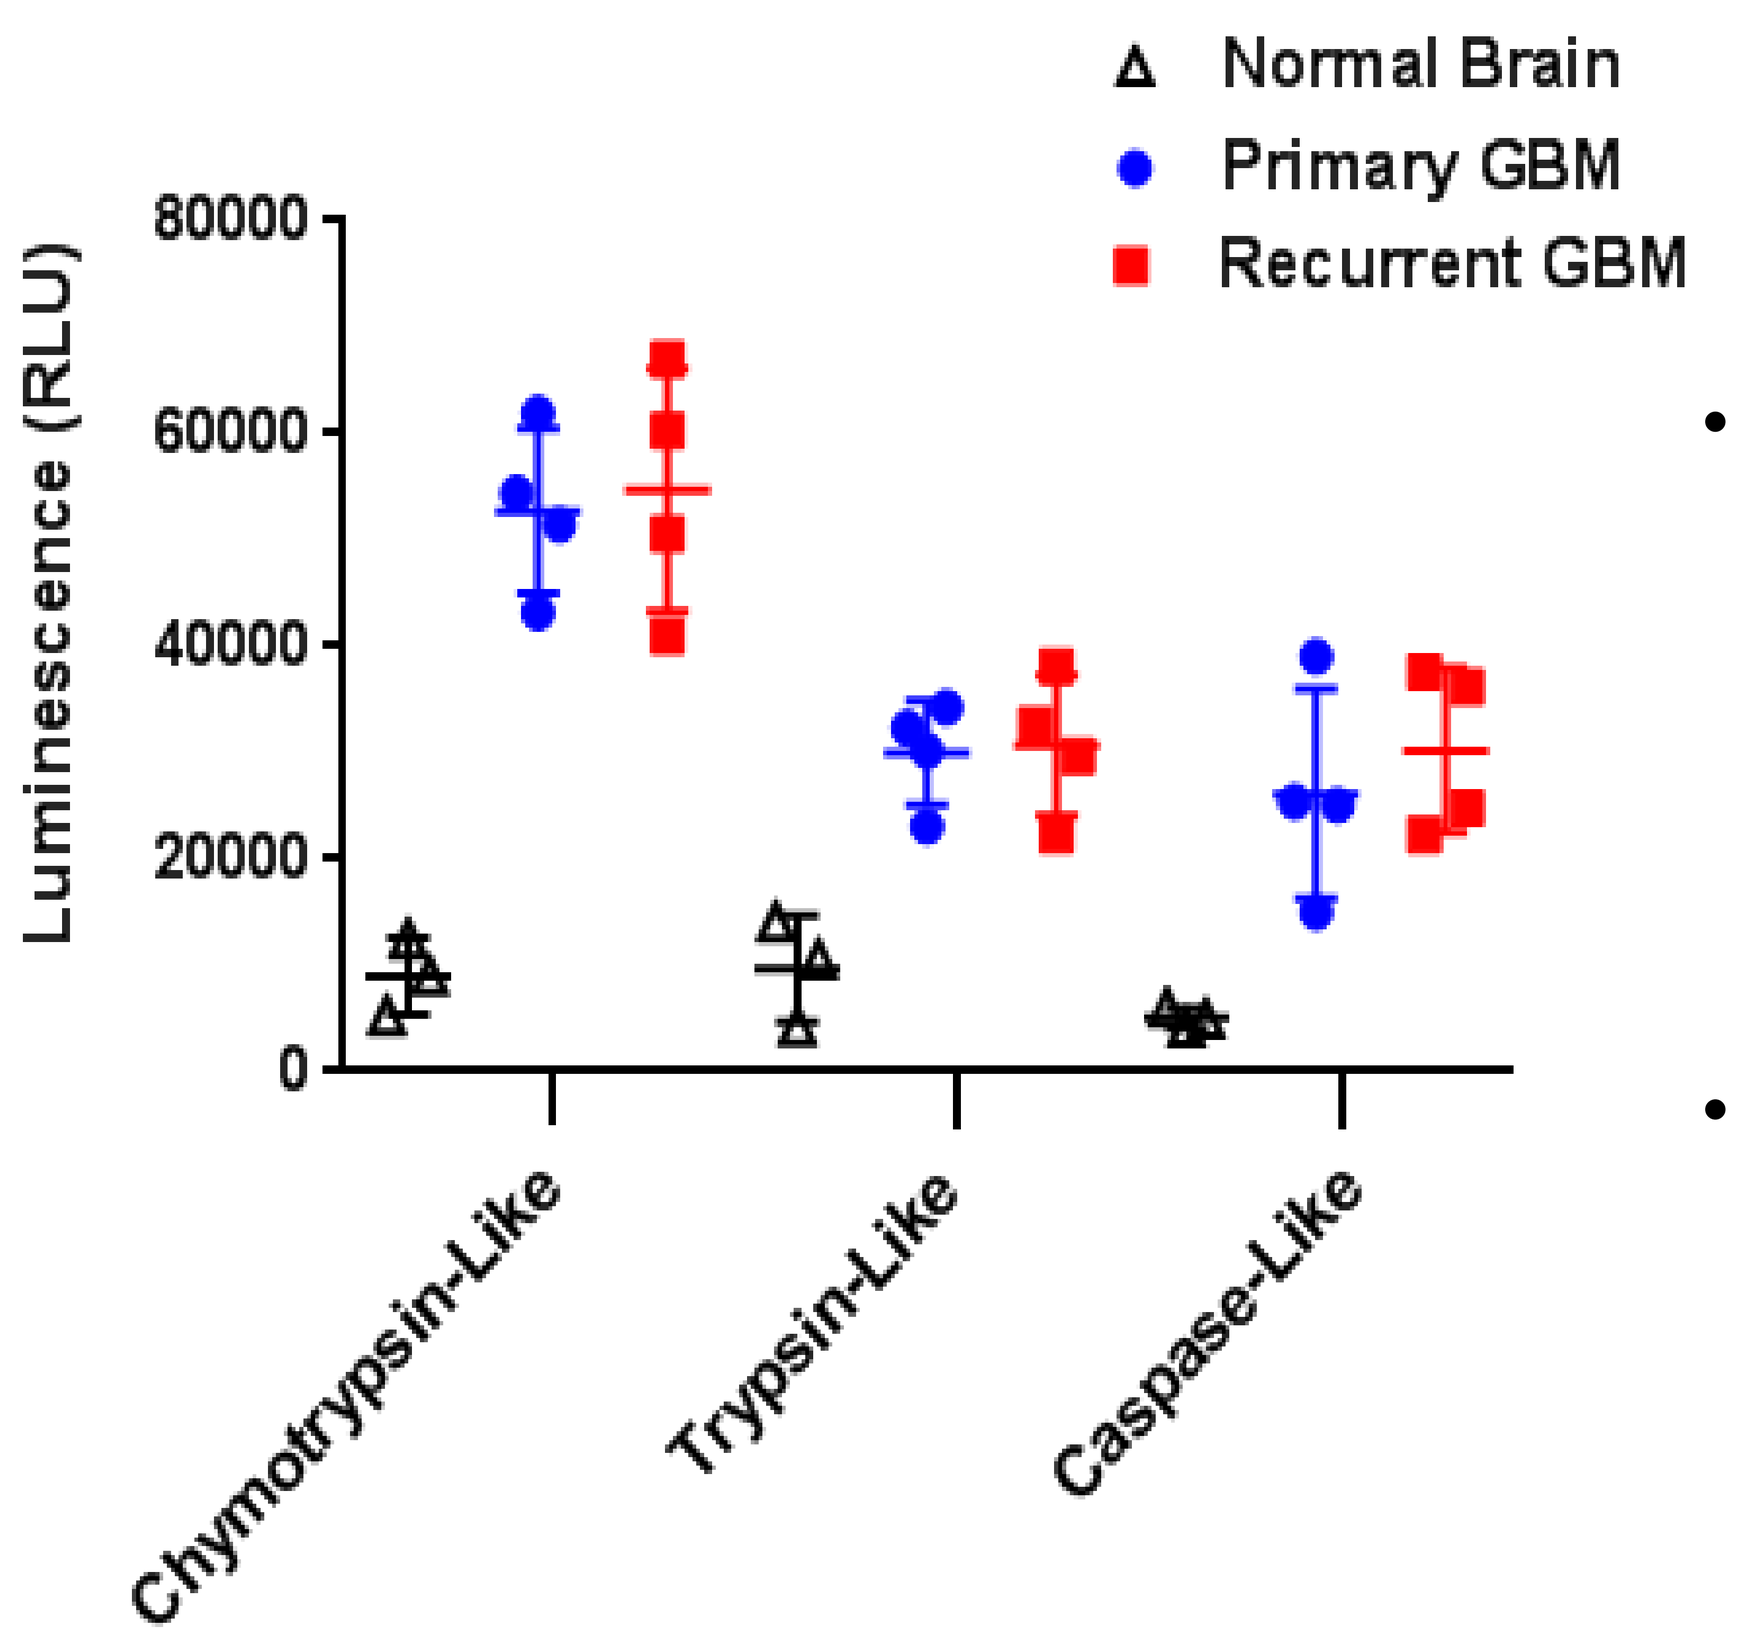
Normal Brain
Primary GBM
Recurrent GBM
Luminescence (RLU)
Chymotrypsin-Like
Trypsin-Like
Caspase-Like

- **Proteasome activities measured in 4 patients with both primary and recurrent glioma and normal brain tissue**
- **All three proteasome activity levels were similarly elevated in both the primary setting and recurrent disease**

Summary & Conclusions

- **In this study we demonstrate that proteasome activity is elevated in glioblastoma patient tumor lysates, and that patients segregate into two groups with high or low proteasome activity**
- **A strong correlation was observed between high CT-L and C-L proteasome activities, suggesting that enzymatic activities within the proteasome may be coordinated and that pan-proteasome inhibition may be necessary to fully inhibit proteasome function in glioma**
- **Proteasome activity was elevated in both primary and recurrent glioblastoma compared to normal brain tissue, suggesting that proteasome inhibition may also have utility as a first-line therapy**
- **Marizomib is a novel irreversible pan-proteasome inhibitor that is currently being evaluated in combination with bevacizumab in grade IV glioma**
- **Future studies will evaluate sensitivity of glioma stem cells (GSCs) to marizomib to determine whether differential proteasome activity can predict response to marizomib in treatment resistant GSCs**